34TH CONGRESS OF THE EUROPEAN COMMITTEE FOR TREATMENT AND RESEARCH IN MULTIPLE SCLEROSIS
10–12 OCTOBER 2018, BERLIN, GERMANY

EP1338

Initial MRI lesion load: a predictor of long-term disability independent of suggestive event

A. Guéguen[1], E. Morvan[1], K. Lecouturier[1], R. Deschamps[1], A. Moulignier[1], A. Bouchareine[1], P. Huillet[2], C. Bensa[1], O. Gout[1].

[1] Service de Neurologie, Fondation Ophtalmologique A. de Rothschild, Paris, France,
[2] Mutual Colour Limited 53-55 Lockhart Road, Hong Kong, China

Objectives

- To asses initial MRI lesion load as a predictor of long-term disability and relation with history of suggestive event.

Methods

- Inclusion criteria were 1) first documented demyelinating event occurred > 14 years ago. 2) initial brain MRI data available.

- Outcome mesure :Time to reach EDSS 3 and 6
- Explicative variables : suggestive event (yes / no) , T2 lesions (n ≥ 14, < 14), gadolinium lesions (n ≥ 2, < 2).
- Number of brain MRI T2 lesions and T1 Gadolinium lesions were determined by two readers. To be confirmed the suggestive event should be supported by clinic and MRI.

- Roc curve were used to define the cuff-off for the number of T2 and T1 Gadolinium lesions. A survival analyse was performed to assess each variable regarding the time to reach EDSS 3 and 6. A cox model was used for multivariable analysis. The correlations between variables were assessed by Spearman test

Results

- Population characteristics are reported in table1 (n = 227)
- The median follow up time is 13.5 years (6,5 - 15.5)

Initial T2 lesions ≥ 14 increases the risk to reach EDSS 3 (figure 2)

- The proportion of patients who reached EDSS 3 was higher in the group with a number of T2 lesions ≥ 14 (0.16, 95%CI(0.10 - 0.25) vs 0.39, 95%CI(0.25 - 0.54); p=0.014) with an increased risk to reach EDSS 3 : HR = 3.03, 95%CI (1.31 - 6,79); p= 0.010
- The number of Gadolinium lesions and the occurence of a suggestive event was not predictive of EDSS 3
- The number of T2 lesions ≥ 14, Gadolinium ≥ 2 and suggestive event was not predictive of EDSS 6

Relation between T2 lesions and the other explicative variables

- Hypothesis : T2 lesion load could be related to the level of disease activity or the duration of presymptomatic phase
- Gadolinium lesions may reflect the disease activity and suggestive event reflects a longer duration of undiagnosed phase
- Number of initial T2 lesions is correlated with Gadolinium lesions (ρ = 0.49, p < 0.001) but not with time elapsed between suggestive event and 1st documented event (ρ = -0.04; p = 0.7)

Conclusion

- Initial MRI T2 lesion load ≥ 14 increases the risk to reach EDSS 3
- Initial MRI T2 lesion load is correlated to G+ lesion

table 1 : Population Characteristics

Age at onset (year)		31.1 (11.8 - 66.4)
Sexe F/M		162/65 (71.4%)
Suggestive event		61 (26.8%)
First event localisation		
	ON	115 (50.6%)
	M	87 (38.3%)
	BS	47 (20.7%)
	H	4 (1.7%)
	MF	20 (9%)
Brain MRI		227 (100%)
Spinal MRI		166 (73.1%)
PL (BOC%)		220 (79.5%)
At the end of Follow up		
CD MS		136 (59.9%)
McD 10 MS		159 (70.4%)
EDSS 3		52 (22.9 %)
EDSS 6		19 (8,3 %)

figure 1 : Initial T2 lesions and CDMS survival

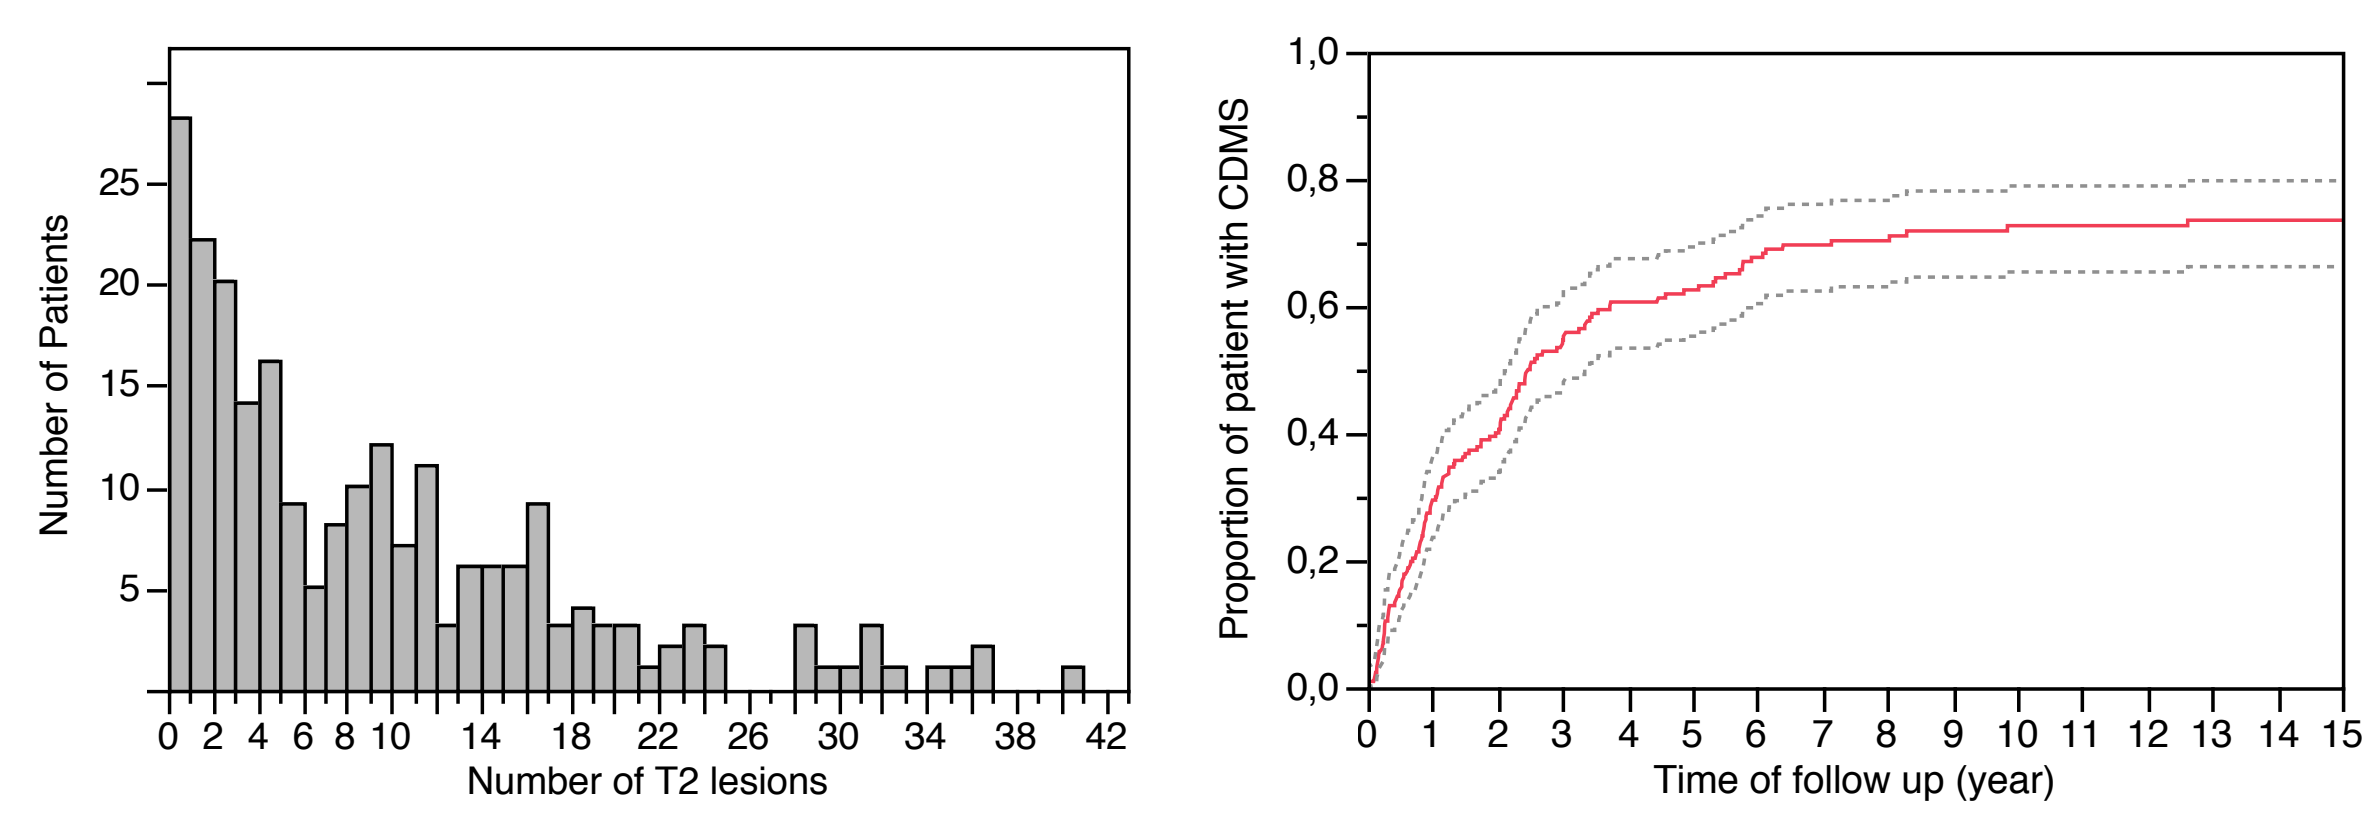
Number of Patients
Number of T2 lesions
Proportion of patient with CDMS
Time of follow up (year)

figure 2 : EDSS 3 Survival Analysis

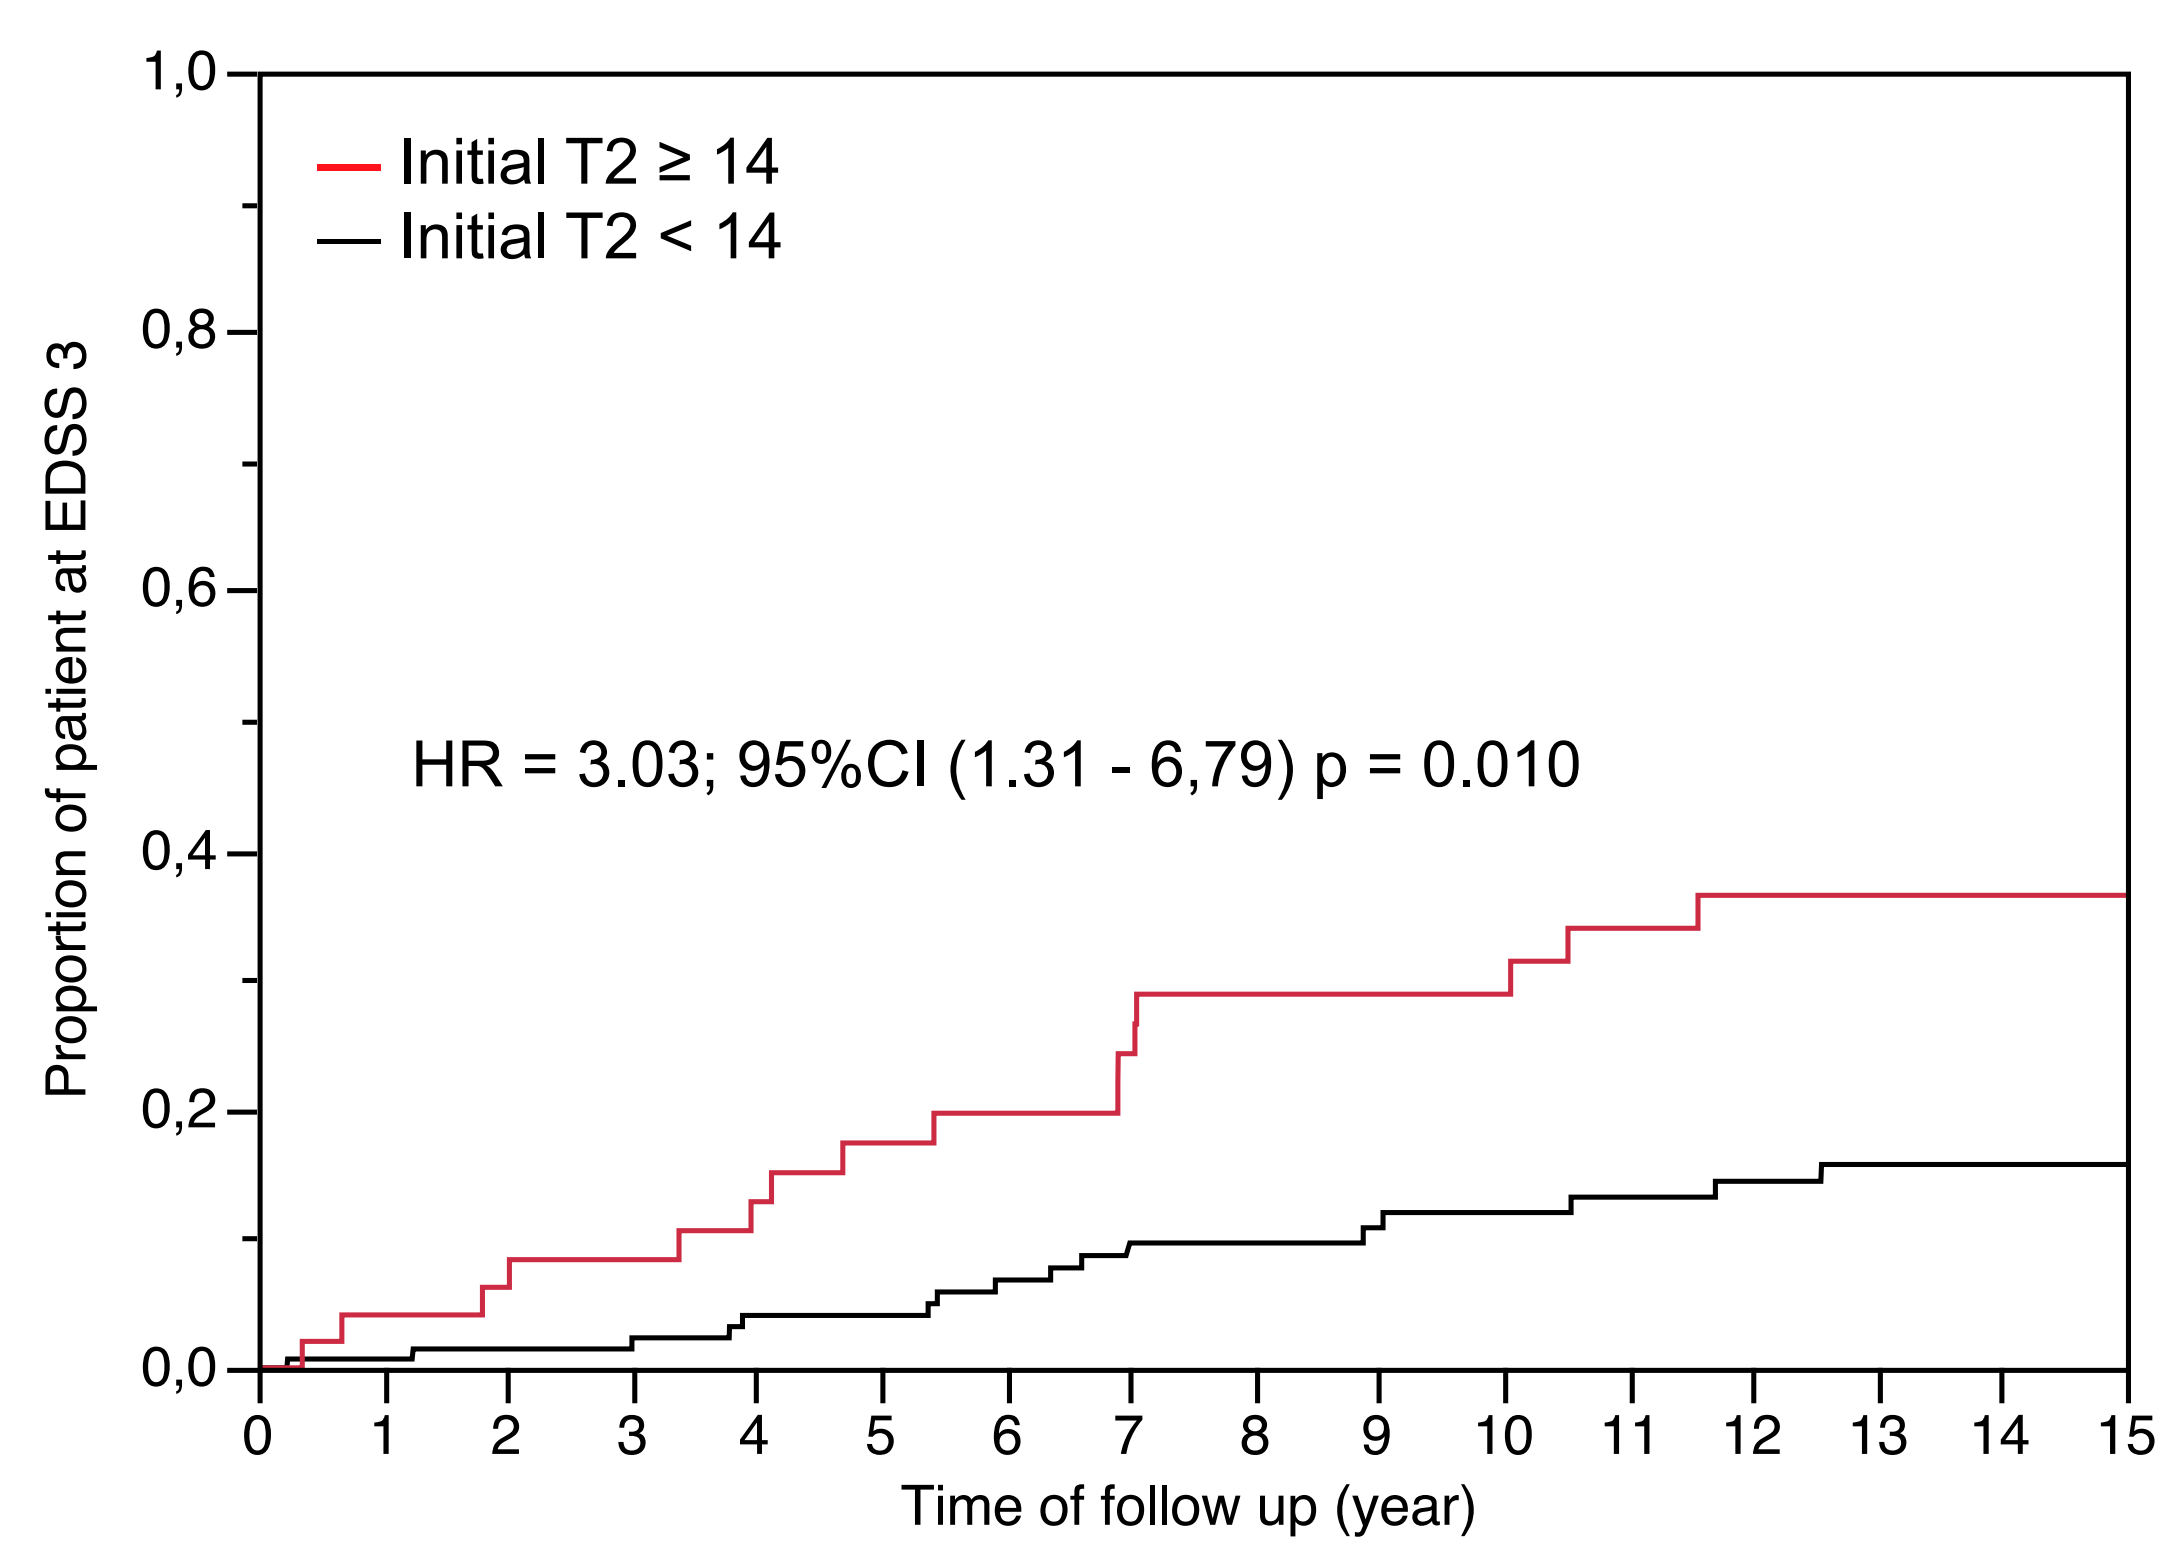
Initial T2 ≥ 14
Initial T2 < 14
HR = 3.03; 95%CI (1.31 - 6,79) p = 0.010
Proportion of patient at EDSS 3
Time of follow up (year)

Mutual colour Ltd.
You'll never walk alone
Fondation A. de Rothschild
De l'œil au cerveau
OFSEP
Observatoire Français de la Sclérose en Plaques